Trofinetide: a Novel Approach to Rett Syndrome

Jeffrey L. Neul,[1] Alan K. Percy,[2] Timothy A. Benke,[3] Elizabeth M. Berry-Kravis,[4] Daniel G. Glaze,[5] Nancy E. Jones,[6] James M. Youakim[7]

[1]Vanderbilt Kennedy Center, Vanderbilt University Medical Center, Nashville, TN, USA; [2]University of Alabama at Birmingham, Birmingham, AL, USA; [3]Children's Hospital of Colorado/University of Colorado School of Medicine, Aurora, CO, USA; [4]Rush University Children's Hospital, Chicago, IL, USA; [5]Texas Children's Hospital/Baylor College of Medicine, Houston, TX, USA; [6]Neuren Pharmaceuticals Limited, Melbourne, Australia; [7]ACADIA Pharmaceuticals Inc., Princeton, NJ, USA

BACKGROUND

- Rett syndrome (RTT), which affects approximately 1 in 10,000 live female births, is a debilitating X-linked neurodevelopmental disorder characterized by apparently normal development for the first 6 months of life, followed by a period of rapid developmental regression and a subsequent plateau period that may persist for decades[1,2]
 - Regression typically occurs between 1 and 4 years of age and is characterized by a failure to reach developmental milestones, loss of expressive language and purposeful hand use, onset of stereotypical hand movements, and other cognitive, motor, and autonomic symptoms[1,2]
 - Precise measurement of the extent of cognitive impairment is difficult because of the severe communication and motor deficits affecting most individuals
 - As a result of these manifestations, RTT confers a substantial burden on affected individuals and their families and caregivers
- There are no approved therapies for RTT; treatment focuses on symptom management
- Trofinetide (glycyl-L-2-methylprolyl-L-glutamic acid) is a novel synthetic analog of 3 amino acids (glycine–proline–glutamate [GPE]), a naturally occurring protein in the brain[3]
 - GPE has been shown to partially reverse core clinical features in an RTT mouse model[3]
- In a recent phase 2 study of female participants with RTT aged 5–15 years, a benefit was observed with trofinetide 200 mg/kg twice daily versus placebo on 3 of 5 measures: the Rett Syndrome Behaviour Questionnaire (RSBQ; *P* = 0.042), the Clinical Global Impressions-Improvement (CGI-I, *P* = 0.029), and the RTT-Clinician Domain Specific Concerns-Visual Analog Scale (*P* = 0.025); trofinetide was well tolerated[4]
- With few RTT-specific outcome measures available for use in clinical trials, identification and use of assessments that address aspects of core features of RTT will enable a more robust evaluation of the clinical effects of interventions

OBJECTIVE

- To present the design of a phase 3 study that utilizes novel scales to investigate the efficacy and safety of trofinetide versus placebo in girls and women with RTT

METHODS

Study Design

- LAVENDER (NCT04181723) is a pivotal, phase 3, double-blind, placebo-controlled trial of trofinetide in females of 5–20 years of age with RTT, designed to investigate its efficacy and safety
- A total of 184 individuals meeting inclusion/exclusion criteria (Table 1) are expected to be randomized, stratified according to age (5–10 years, 11–15 years, and 16–20 years) and baseline RSBQ severity (total score of <35 and ≥35) to receive 12 weeks of trofinetide or placebo (Figure 1)
- This study is followed by a 40-week open-label extension study

Figure 1. LAVENDER study design

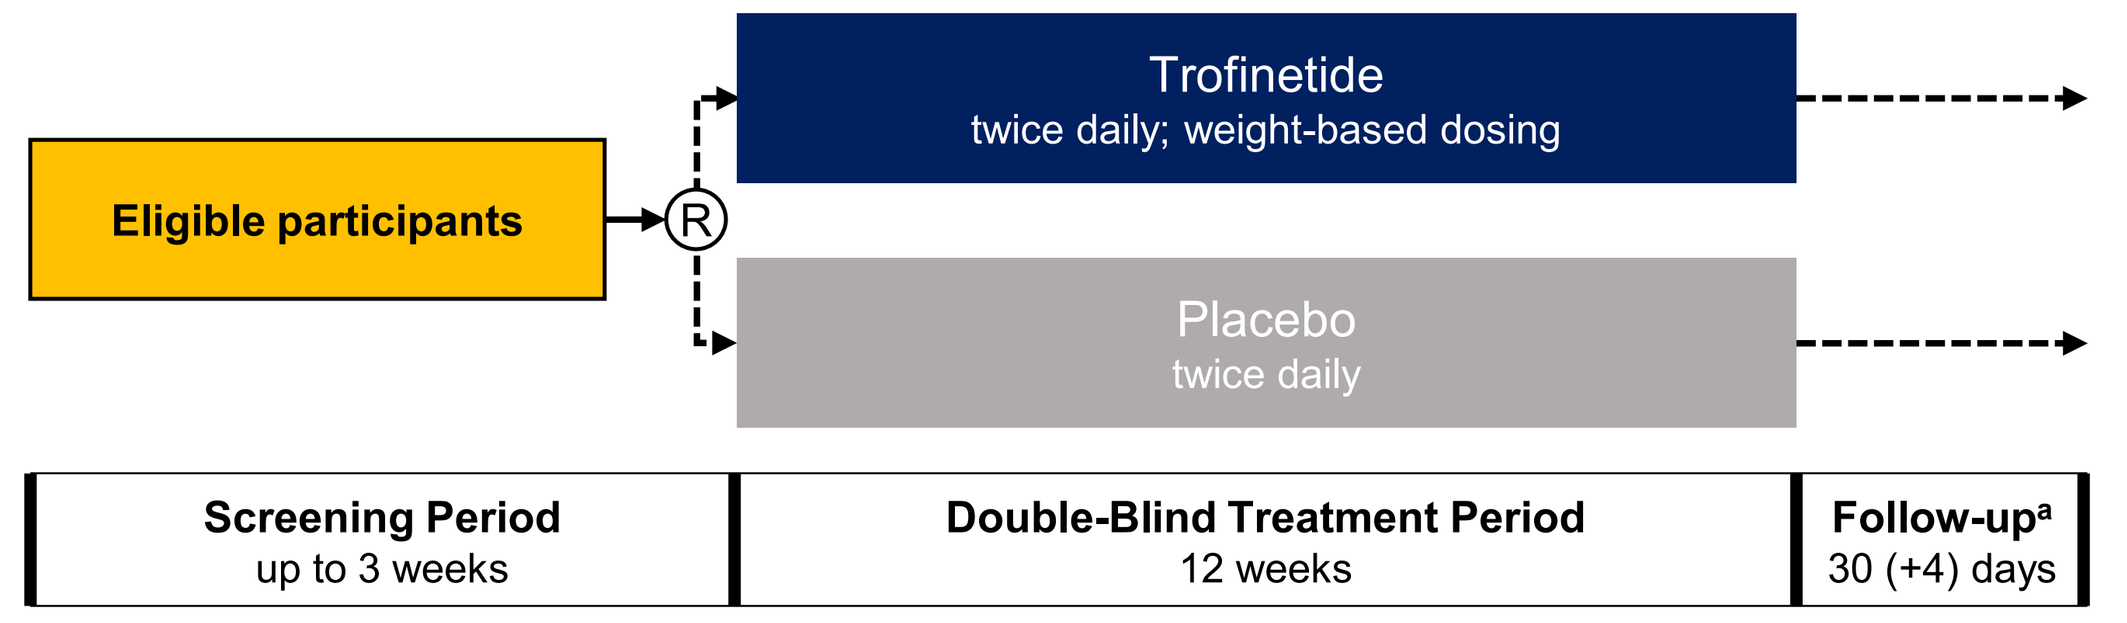
Trofinetide
twice daily; weight-based dosing
Eligible participants
R
Placebo
twice daily
Screening Period
up to 3 weeks
Double-Blind Treatment Period
12 weeks
Follow-up[a]
30 (+4) days

[a]If the individual continues into the OLE from the current study, she will not complete the follow-up visit and will roll over into the OLE
OLE, open-label extension

Table 1. LAVENDER key inclusion and exclusion criteria

Inclusion criteria	Exclusion criteria
• Girls and women, 5–20 years of age	• Current clinically significant cardiovascular, endocrine (such as hypo- or hyperthyroidism, type 1 diabetes, or uncontrolled type 2 diabetes), renal, hepatic, respiratory, or gastrointestinal disease (such as celiac disease or inflammatory bowel disease), or major surgery planned during the study
• Weight ≥12 kg	• Treated with insulin within 12 weeks of baseline
• Classic/typical RTT	• Known history or symptoms of long QT syndrome
• Documented disease-causing mutation in *MECP2* gene	• QTcF interval >450 ms
• Stable pattern of seizures or no seizures within 8 weeks of screening	• Treatment with insulin, IGF-1, or growth hormone within 12 weeks of baseline
• RTT Clinical Severity Scale rating of 10–36	
• CGI-S score of ≥4	

CGI-S, Clinical Global Impressions-Severity; IGF-1, insulin-like growth factor 1; QTcF, corrected QT interval using Fridericia's correction; RTT, Rett syndrome

Efficacy Assessments

- Efficacy endpoints will evaluate change from baseline to Week 12 using established and novel efficacy assessments for RTT

Co-Primary Endpoints

- The Rett Syndrome Behaviour Questionnaire (RSBQ)
 - The RSBQ is a 45-item rating scale completed by the caregiver that assesses a range of neurobehavioral features impaired in RTT[5]
 - The caregiver rates items as "0" (not true), "1" (somewhat or sometimes true), or "2" (very true)
 - This instrument is well validated and used widely to assess RTT symptoms in a number of RTT studies,[6-12] including the phase 2 trofinetide trial[4]
 - The RSBQ has been correlated with functioning and quality of life and validated across a range of ages (2–47 years) in RTT[13-15]
- The Clinical Global Impression–Improvement (CGI-I)
 - The clinician rates how much the affected individual's illness (RTT as a whole) has improved or worsened versus baseline on a 7-point scale[16]
 - CGI-I ratings will be assessed using RTT-specific anchors[17] across major symptom areas (Table 2) as in previous trofinetide and other studies[4,17,18]
 - An effort was made to standardize rating of the CGI-I and CGI-Severity (CGI-S) across all of the study sites
 - An expert panel wrote a series of clinical case vignettes
 - All CGI raters were trained on the panel's "gold standard" CGI-I and CGI-S ratings for each vignette before beginning to rate in the study

Table 2. Anchors for the Clinical Global Impression Scale-Improvement[17]

Score	CGI-I descriptor	RTT-specific anchors
1	*Very much improved*	• Marked improvement, across settings and/or multiple behavior problems • Improvement must be substantial; usually accompanied by considerable caregiver enthusiasm; usually noticeable behavioral improvement in the clinic • A CGI-I of 1 does not require a CGI-S rating better than baseline; usually the CGI-S does also improve
2	*Much improved*	• Moderate improvement in a single symptom area (especially if seen across settings) or moderate improvements in several areas (even if confined to one setting), characterized by durability. For example, a change reported for a few days is unlikely to warrant such a rating, but one that coincided with treatment and was evident for at least the last week would likely warrant a rating of 2 • A CGI-S rating better than baseline is not required to receive a CGI-I rating of 2, but often (not always) the CGI-S also improves
3	*Minimally improved*	• Modest improvements, especially if confined to one setting. Trivial changes or changes that are possibly present or require guesswork usually would be scored as 4 (the level below this one)
4	*No change*	• The absence of change in behavior or clinical presentation from baseline. Chance fluctuations and equivocal improvements or declines should be included here
5	*Minimally worse*	• Some worsening in symptoms that are mild to moderate or may be confined to one setting
6	*Much worse*	• Moderate to moderately severe worsening, including in a single symptom area when observed across settings. Moderately severe changes that are confined to one setting may warrant a rating of "Much worse"
7	*Very much worse*	• Significant worsening, across settings and/or across multiple symptoms

CGI-Improvement is a rating of change; normalization is not necessary for a rating of 1, although if behavior is normalized it suggests a CGI-I of 1
A CGI-I of 2 is appropriate for definite, unequivocal improvement of a magnitude that makes the clinician confident that the treatment is helping
A score of 3 (or 5) is appropriate if variations in ratings and other criteria appear to represent more than random chance or rating error, but are moderate in scope
A score of 4 is appropriate for slight variation in either direction of a magnitude that is likely due to chance, natural history, external events, or rating error
CGI-I, Clinical Global Impressions-Improvement; CGI-S, Clinical Global Impression-Severity

Secondary Endpoints

- In addition to improvement in the symptoms of RTT overall, assessed by the RSBQ, CGI-I, and CGI-S, the study team also wanted to assess:
 - Symptoms and functioning domains of key concern in this population; particularly, communication, ambulation, and use of hands
 - Quality of life
 - Caregiver burden
- Ideally, measures would be RTT-specific or had been shown to detect change in an RTT population
 - Stand-alone measures were preferred over subscales
 - A balance of caregiver- and clinician-completed assessments was preferred

Key Secondary Endpoint

- The Social Composite score from the Communication and Symbolic Behavior Scales Developmental Profile™ Infant-Toddler Checklist (CSBS-DP-IT Social) is used as the key secondary endpoint
 - Improvement in the ability to communicate is one of the most important goals for caregivers in the treatment of RTT
 - The CSBS-DP-IT Checklist has shown evidence of sensitivity to change in behavioral intervention studies in other developmental disorders[19,20]
 - CSBS-DP-IT Social and items from the CSBS-DP-IT Checklist have been used in at least 2 prior studies of girls and women with RTT[7,21]
 - The CSBS-DP-IT Checklist is 1 of 3 components of the CSBS-DP, which was developed to assess communication and pre-linguistic skills in young children (12–24 months of age)[19] and can be used with older children with developmental delay[20,21]
 - The CSBS-DP-IT Checklist is a 24-item rating scale completed by the caregiver, with each item scored using a 3-level rating of frequency: "not yet", "sometimes", and "often"
 - The Checklist is composed of 3 composite scores: Social, Speech, and Symbolic
 - The first 13 items make up the Social composite score
 - The checklist and scoring card can be accessed online[22]

Other Secondary Endpoints

- Other secondary endpoints will evaluate the impact of the disease on the daily life of the individual with RTT and of the family, the quality of life of the individual with RTT, the toll that dealing with RTT takes on the caregiver, and overall disease severity (Table 3)
 - Following best practice, CGI-S ratings will employ RTT-specific anchors across major symptom areas[17]
- The Impact of Childhood Neurologic Disability Scale (ICND) total score and Overall Quality of Life Rating of the ICND
 - The ICND was developed to evaluate the impact that a child's condition has on the child's and the family's everyday life at the present time and during the previous 3 months[23]
 - The parent or other caregiver evaluates the effect of 4 conditions or health problems on 11 aspects of the child's or the family's life as "A lot", "Some", "A little", "Not at all", or "Does not apply"
 - The caregiver then rates overall quality of life of the subject by responding to the following: "Please rate your child's overall 'Quality of Life' on the scale below. Choose the number which you feel is best and circle it". The choices range from 1 ("Poor") to 6 ("Excellent")
- The Rett Syndrome Caregiver Burden Inventory (RTT-CBI)
 - The RTT-CBI (or Rett Caregiver Inventory Assessment [RTT-CIA]) is a syndrome-specific, caregiver-completed questionnaire that is based on the CBI designed for Alzheimer's disease[24,25]
 - The RTT-CBI has been used in the RTT Natural History study[24] and the phase 2 trofinetide studies[4,18]
- Clinical Global Impression-Severity (CGI-S)
 - The CGI-S will employ RTT-specific anchors[17]; this scale was used in the phase 2 trofinetide studies and is being used in other clinical studies

Table 3. Other secondary endpoints

Assessment	Area evaluated	Method of response
Impact of Childhood Neurologic Disability Scale (ICND) Total Score[a]	Impact that a child's condition has on the child's and the family's everyday life at the time of assessment and during the previous 3 months[23] Four conditions are assessed: 1. Inattentiveness, impulsivity, or mood 2. Ability to think and remember 3. Neurologic or physical limitations 4. Epilepsy	Parent or caregiver rates the effect of the 4 conditions/health problems on 11 aspects of the child's or the family's life as "A lot", "Some", "A little", "Not at all", or "Does not apply"
Overall Quality of Life Rating of the ICND	Overall quality of life of the affected individual	Caregiver response to: "Please rate your child's overall 'Quality of Life' by selecting the number they feel is best on a scale ranging from 1 ("Poor") to 6 ("Excellent")
Caregiver Burden Inventory (RTT-CBI) Total Score (items 1–24)	Caregiver burden (directly) and significance of treatment on activities of daily living (indirectly)	Caregivers rate frequency each statement describes their feeling or experience on a 5-point Likert scale: 0-never; 1-rarely; 2-sometimes; 3-frequently, and 4-nearly always Items 1–24 are negatively worded, with a total maximal score of 96[19] Consistent with previous studies of trofinetide,[4,18] the total score is defined as the Total Burden Score
Clinical Global Impression-Severity (CGI-S)	Severity of the participant's illness (RTT as a whole)	Clinicians rate illness severity relative to their experience with others with the same diagnosis

[a]The scale can be accessed online (see Appendix I)[23]
CGI-S, Clinical Global Impression-Severity; RTT, Rett syndrome

- Four novel RTT-specific clinical rating scales, which evolved from the RTT-Clinician Domain Specific Concerns—Visual Analog Scale (RTT-DSC-VAS) used in the trofinetide phase 2 study,[4] will assess core symptoms of RTT (Table 4)
 - For these scales, clinicians rate participant ability on an 8-point Likert scale, from 0 (normal function) to 7 (most severe impairment)

Table 4. Novel RTT-specific clinical rating scales used for secondary outcomes[a]

		Hand Function (RTT-HF)©	Ambulation and Gross Motor Skills (RTT-AMB)©	Ability to Communicate Choices (RTT-COMC)©	Verbal Communication (RTT-VCOM)©
Area evaluated		Ability to use hands for functional purposes (eg, reaching for and grasping objects, self-feeding, drawing)	Ability to sit, stand, and ambulate (eg, walking, running, climbing stairs)	Ability to communicate choices or preferences, including nonverbal means such as eye contact or gestures	Ability to communicate verbally (eg, words and phrases)
Score rating	0	Normal, no impairment	Normal, no impairment	Normal	Normal, no impairment
	1	Can grip a writing instrument effectively and can draw a shape (or has an equivalent fine motor skill) BUT Still has observable fine motor problems	Stands and sits independently AND Can move from sit to stand AND Walks independently AND Can climb up and down stairs or run May still have evidence of dystonia, ataxia, or dyspraxia	Makes a forced choice between 2 or more drawings or symbolic representations	Uses phrases (not exclusively "fixed phrase") or sentences *May still feature echolalia or perseveration*
	2	Can grasp objects AND Can self-feed AND Has a pincer grasp	Stands and sits independently AND Can move from sit to stand AND Walks independently	Makes a forced choice between 4 photographs of objects	Uses many words (>20), which may include short "fixed phrases" that are used as if they were a single word *May feature echolalia or perseveration*
	3	Can grasp objects AND Can self-feed BUT Does not have a pincer grasp	Stands and sits independently >30 seconds AND Can walk independently but only a short distance and with reduced pace	Makes a forced choice between 2 photographs of objects	Uses a few words (5–20) AND The words are generally context-appropriate
	4	Reaches for and can grasp an object BUT Cannot self-feed (ie, the hand functioning necessary to feed oneself or drink by oneself)	Stands and sits independently >30 seconds AND Walks with support across a range of distances	Makes a forced choice between 2 real-life items (eg, food, toys, videos, shapes)	Uses <5 words AND The words are not necessarily context-appropriate *Often occurs when stressed, anxious or uncomfortable*
	5	Regularly reaches for objects AND Can hold object >2 seconds if placed in the hand BUT Cannot grasp objects	Stands AND Sits independently >30 seconds OR Walks only a short distance (such as a few meters) with support	Makes unforced choices (eg, chooses food, a toy, a video)	No words BUT Babbles (makes consonant-vowel combination sounds)
	6	Rarely or only occasionally reaches for an object OR Can hold object >2 seconds if placed in the hand BUT Cannot grasp objects	Sits independently >30 seconds OR Stands with support but unable to take steps	Responds to name by looking at speaker; Does not make choices	No words AND Makes vocalizations (vowel-only sounds) BUT Does not babble (ie, does not make consonant-vowel combination sounds)
	7	No hand use	Cannot sit without support AND Cannot stand AND Cannot walk	No interactions or no attempts to respond to requests even from caregivers; Does not make choices	No words AND No vocalizations (may scream)

[a]Further development of scales used in Study Neu-2566-RETT-002,[4] including the RTT-DSC Hand Use Rating, RTT-DSC Ambulation Rating, and RTT-DSC Language/Communication Rating scales
DSC, Clinician Domain Specific Concerns; RTT, Rett syndrome
© 2019 ACADIA Pharmaceuticals, Inc.

Safety Endpoints

- Treatment-emergent adverse events
- Serious adverse events
- Withdrawals due to adverse events
- Potentially clinically important changes in other safety assessments

Pharmacokinetic (PK) Endpoints

- Whole blood concentration of trofinetide and possible metabolites
- Trofinetide PK parameters using the population PK approach
- PK/pharmacodynamics (PD) using appropriate PK/PD analysis methods

Assessment Schedule

- Assessments will be made at time points indicated in Figure 2

Figure 2. Summary of timing of assessments and assessors in LAVENDER

	Timing of assessment		Treatment period, Week			Assessed by:	
	SC	BL	2	6	12	Caregiver	Clinician
Co-primary endpoints							
RSBQ	■	■	■	■	■	■	
CGI-I			■	■	■		■
Key secondary endpoint							
CSBS-DP-IT		■	■	■	■	■	
Other secondary endpoints							
CGI-S	■	■	■	■	■		■
ICND including QoL		■			■	■	
RTT-CBI		■			■	■	
RTT-HF		■	■	■	■		■
RTT-AMB		■	■	■	■		■
RTT-COMC		■	■	■	■		■
RTT-VCOM		■	■	■	■		■

AMB, Ambulation and Gross Motor Skills; BL, baseline; CBI, Caregiver Burden Inventory; CGI-I, Clinical Global Impressions-Improvement; CGI-S, Clinical Global Impressions-Severity; COMC, Ability to Communicate Choices; CSBS-DP-IT, Communication and Symbolic Behavior Scales Developmental Profile™ Infant-Toddler Checklist; HF, Hand Function; ICND, Impact of Childhood Neurologic Disability Scale; QoL, quality of life; RSBQ, Rett Syndrome Behaviour Questionnaire; SC, screening; VCOM, Verbal Communication

CONCLUSIONS

- Significant unmet needs remain in the treatment of individuals with RTT
- Established measures, as well as novel disease-specific scales optimized to demonstrate change in core clinical features of RTT, are critical to demonstrate the effects of therapeutic interventions in clinical trials of individuals with RTT
- The data from this phase 3 clinical trial, which includes RTT-specific outcome assessments, may support the use of trofinetide as an effective, safe, and tolerable treatment for RTT

REFERENCES

1. Neul JL, et al. *Ann Neurol.* 2010;68(6):944-950.
2. Hagberg B. *Ment Retard Dev Disabil Res Rev.* 2002;8(2):61-65.
3. Tropea D, et al. *Proc Natl Acad Sci U S A.* 2009;106(6):2029-2034.
4. Glaze DG, et al. *Neurology.* 2019;92(16):e1912-e1925.
5. Mount RH, et al. *J Child Psychol Psychiatry.* 2002;43(8):1099-1110.
6. Khwaja OS, et al. *Proc Natl Acad Sci U S A.* 2014;111(12):4596-4601.
7. O'Leary HM, et al. *Ann Clin Transl Neurol.* 2018;5(3):323-332.
8. Anagnostou E. https://clinicaltrials.gov/ct2/show/NCT04041713. Accessed April 28, 2020.
9. Anavex Life Sciences Corp. https://clinicaltrials.gov/ct2/show/NCT04304482. Accessed April 28, 2020.
10. Anavex Life Sciences Corp. https://clinicaltrials.gov/ct2/show/NCT03941444. Accessed April 28, 2020.
11. Anavex Life Sciences Corp. https://clinicaltrials.gov/ct2/show/NCT03758924. Accessed April 28, 2020.
12. GW Research Ltd. https://clinicaltrials.gov/ct2/show/NCT03848832. Accessed April 28, 2020.
13. Barnes KV, et al. *J Neurodev Disord.* 2015;7(1):30.
14. Cianfaglione R, et al. *J Neurodev Disord.* 2015;7(1):11.
15. Robertson L, et al. *Am J Med Genet B Neuropsychiatr Genet.* 2006;141B(2):177-183.
16. Busner J, Targum SD. *Psychiatry (Edgmont).* 2007;4(7):28-37.
17. Neul JL, et al. *J Child Neurol.* 2015;30(13):1743-1748.
18. Glaze DG, et al. *Pediatr Neurol.* 2017;76:37-46.
19. Wetherby AM, et al. *J Speech Lang Hear Res.* 2002;45(6):1202-1218.
20. Anagnostou E, et al. *Autism.* 2015;19(5):622-636.
21. Urbanowicz A, et al. *J Speech Lang Hear Res.* 2016;59(6):1373-1383.
22. CSBS DP IT Checklist. https://brookespublishing.com/wp-content/uploads/2012/06/csbs-dp-itc.pdf. Accessed April 28, 2020.
23. Camfield C, et al. *Dev Med Child Neurol.* 2003;45(3):152-159. https://onlinelibrary.wiley.com/doi/epdf/10.1111/j.1469-8749.2003.tb00923.x. Accessed April 28, 2020.
24. Lane JB, et al. *J Autism Dev Disord.* 2017;47(4):1102-1112.
25. Novak M, Guest C. *Gerontologist.* 1989;29(6):798-803.

ACKNOWLEDGMENTS

The study was supported by ACADIA Pharmaceuticals Inc. (San Diego, CA, USA). The authors thank Josh Corriveau, PharmD, MBA of Scientific Commercialization LLC for contributions to abstract preparation and coordination, which was funded by ACADIA Pharmaceuticals Inc. Medical writing support was provided by Beth Burke, PhD, CMPP, and Lynne Isbell, PhD, CMPP, of Evidence Scientific Solutions, Inc. and funded by ACADIA Pharmaceuticals Inc.

DISCLOSURES

JLN has received personal compensation for consulting services from ACADIA Pharmaceuticals Inc., AveXis, GW Pharmaceuticals, and Paraxel. He is a member of a data safety monitoring board for a clinical trial conducted by Ovid Therapeutics. **AKP** is co-editor of *Translational Science of Rare Diseases*. **TAB** received research funding from International Foundation for CDKL5 Research, Loulou Foundation, and National Institutes of Health; consultancy for AveXis, GW Pharmaceuticals, International Rett Syndrome Foundation, Marinus, Ovid, and Takeda; clinical trials with ACADIA, GW Pharmaceuticals, Marinus, Ovid, and RSRT; All remuneration has been made to his department. **EMB-K** has received funding from ACADIA Pharmaceuticals, BioMarin, Cydan, E-Scape Bio, Fulcrum, GeneTx, GW Pharmaceuticals, Healyx, Ionis, Lumos Pharma, Mallinckrodt, Orphazyme, Ovid, RegenxBio, Roche, Ultragenyx, Yamo, and Zynerba to consult on trial design or development strategies in fragile X syndrome or other neurodegenerative disorders. All funding to EMB-K is directed to Rush University Medical Center (RUMC) in support of rare disease programs; EMB-K receives no personal funds. EMB-K has received research support to RUMC from GW Pharmaceuticals, Ionis/Biogen, Lumos Pharma, Mallinckrodt, Newron Pharmaceuticals, Ovid, Retrophin, Roche, Tetra, Ultragenyx, and Zynerba to perform clinical trials in neurodevelopmental and neurodegenerative disorders, and from Asuragen to develop testing standards for FMR1 testing and validate FMR1 and SMN assays. **DGG** has received personal compensation and research support from ACADIA Pharmaceuticals, Neuren Pharmaceuticals, and Newron Pharmaceuticals. **JMY** is an employee of and stockholder in ACADIA Pharmaceuticals Inc. **NEJ** is an executive at Neuren Pharmaceuticals, Ltd.

To receive a copy of this poster, scan QR code via barcode reader application.

By requesting this content, you agree to receive a one-time communication using automated technology. Message and data rates may apply. Links are valid for 30 days after the congress presentation.

P14.004

American Academy of Neurology 72nd Annual Meeting 2020

ACADIA® Pharmaceuticals